AWCA & 7th GLOBAL WOUND CONFERENCE 2025

MALAYSIA

ACCELERATED HEALING OF A CHRONIC WOUND IN AN UNCONTROLLED DIABETIC PATIENT USING A BIOMIMETIC SCAFFOLD LOADED WITH MSC-DERIVED EXOSOMES: A CASE REPORT

Syafiqah Mohd Satali[1], Hamishatul Shamsiah Abdul Manaf[1], Nurul Hamizah Binti Amran[1], Norhasliza Zaina Abidin[1], Shukur Ahmad[2]

[1]Wound Care Unit, Hospital Teluk Intan

[2]Orthopedic Department, Hospital Teluk Intan

INTRODUCTION

Chronic wounds are a major health burden, particularly in patients with uncontrolled diabetes, where impaired angiogenesis, prolonged inflammation, neuropathy, and poor glycaemic control disrupt normal healing(1). Conventional dressings often fail to provide an optimal regenerative environment, resulting in persistent wounds and complications. Mesenchymal stem cells (MSCs) have been studied for their regenerative effects; however, direct stem cell therapy carries challenges such as poor survival, immune rejection, and tumorigenic potential(1).

Emerging evidence suggests that the therapeutic benefits of MSCs are largely mediated by their secreted exosomes, which promote angiogenesis, modulate inflammation, and accelerate tissue repair(1). Despite their promise, MSC-derived exosomes have a short half-life and are rapidly cleared from the wound site. Biomimetic scaffolds, designed to mimic the extracellular matrix, enhance exosome stability, prolong release, and create a pro-healing microenvironment(2). Here, we present a case of a chronic diabetic leg wound treated with a biomimetic scaffold loaded with MSC-derived exosomes, demonstrating accelerated healing and significant wound size reduction.

CASE PRESENTATION

A 62-year-old female with uncontrolled type 2 diabetes mellitus and a 5-year history of chronic non-healing wound over the left leg, secondary to a previous abscess, was selected for this case. Her most recent HbA1c was 8.7%, reflecting poor glycaemic control. She was overweight and a non-smoker. The wound failed to heal despite multiple courses of standard wound care. Due to the chronicity of the wound, uncontrolled diabetes, and poor response to conventional care, she was selected for treatment with a biomimetic scaffold loaded with MSC-derived exosomes.

RESULTS

At initial assessment (15 April 2025), the wound measured 5.0 × 3.5 cm (17.5 cm²) over left leg (Fig. 1.0 and Fig 1.1). Following regular scaffold and exosome applications, progressive improvement was observed with healthy granulation tissue and epithelialisation.

After 11 weeks (1 July 2025), the wound size had reduced to 2.5 × 1.8 cm (4.5 cm²), representing a 74.3% reduction in area (Fig. 2.0 and Fig 2.1). No signs of local infection, inflammation, or adverse reactions were observed during the entire treatment period. The patient tolerated the procedure well, and wound healing continued to progress satisfactorily.

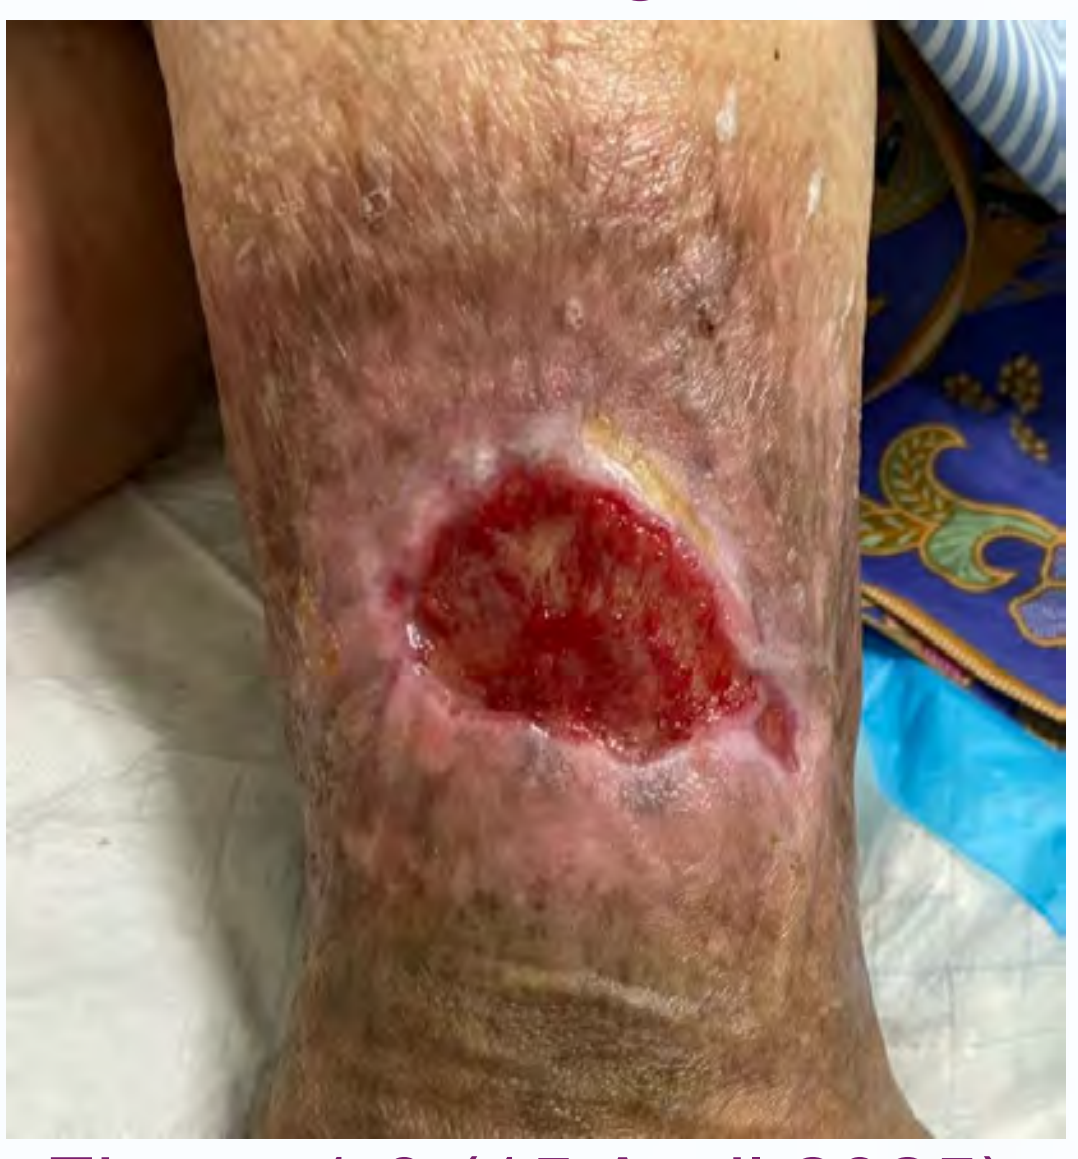

Figure 1.0 (15 April 2025)

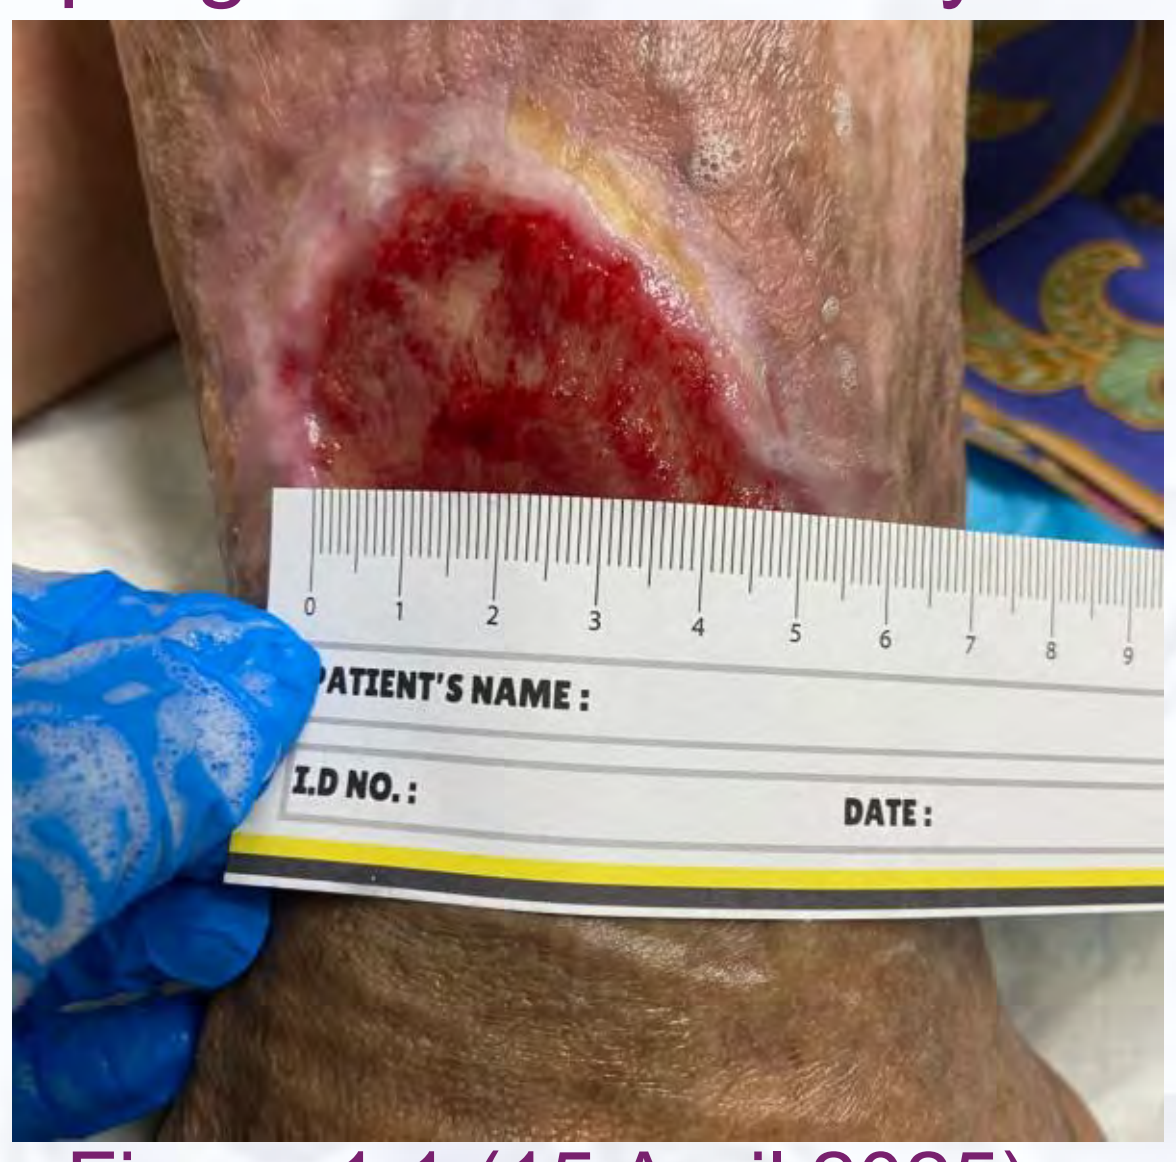
0 1 2 3 4 5 6 7 8 9
ATIENT'S NAME :
I.D NO. :
DATE :

Figure 1.1 (15 April 2025)

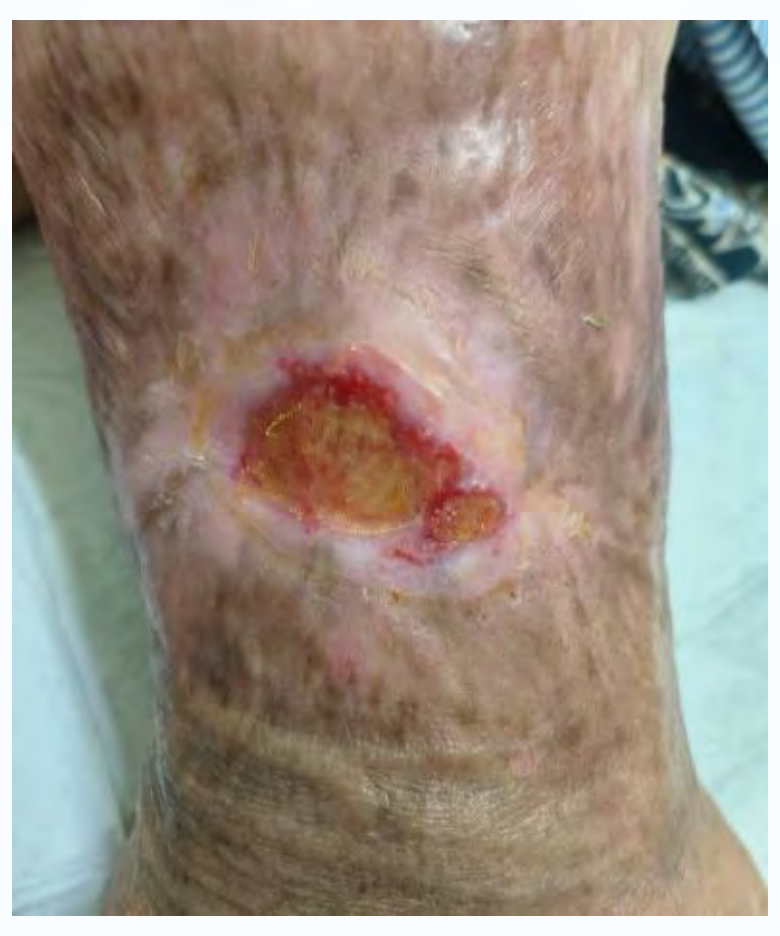

Figure 2.0 (1 July 2025)

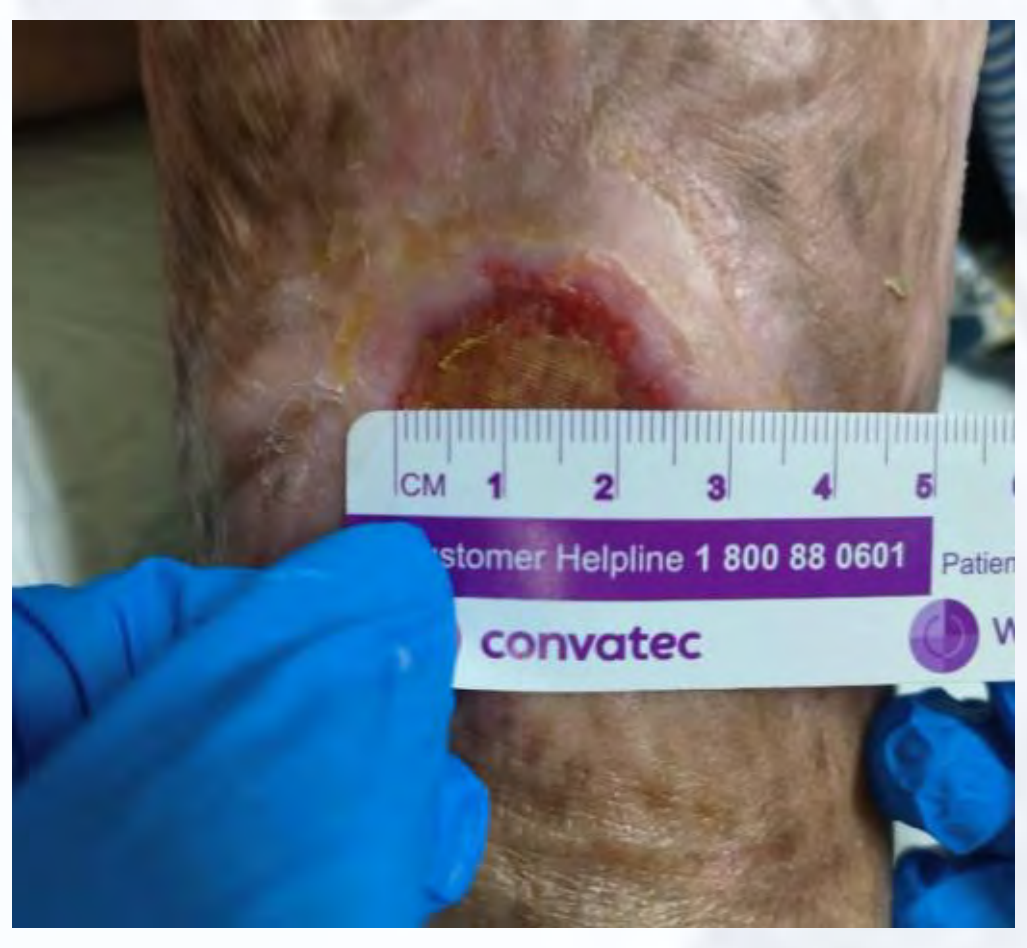
CM 1 2 3 4 5
stomer Helpline 1 800 88 0601
convatec

Figure 2.1 (1 July 2025)

METHODOLOGY

Prior to scaffold application, the wound was soaked, desloughed, and cleansed with an antimicrobial solution. The scaffold was cut to wound size, loaded with exosomes, and applied, followed by 0.3 ml exosome solution. The exosome was allowed to fully absorb before the wound was covered with sterile gauze. Wound assessment was documented at each visit, including wound bed appearance, exudate level, wound size, peri-wound condition, wound pain, and wound management. Twice-weekly reviews at 4–5 days intervals involved soaking, scaffold assessment, trimming, and reloading with 0.3 ml exosomes until healing progressed. Informed consent for treatment and publication was obtained from the patient prior to initiation of therapy.

DISCUSSION

Chronic wounds in diabetic patients are widely acknowledged to be challenging to heal due to impaired angiogenesis, prolonged inflammation, neuropathy, and poor glycaemic control. These pathophysiological changes disrupt the normal healing cascade, making conventional dressings and standard wound care often inadequate in promoting complete recovery(1).

Mesenchymal stem cell (MSC)-derived exosomes have emerged as a promising therapeutic option in regenerative medicine. Exosomes carry bioactive molecules such as proteins, lipids, and microRNAs that promote angiogenesis, stimulate fibroblast proliferation, and modulate inflammation, thereby accelerating tissue regeneration(2)(3). Unlike direct stem cell therapy, exosomes provide these benefits without the risks of immune rejection or tumorigenicity, making them a safer and more practical alternative for clinical use. However, their therapeutic application is limited by their short half-life and rapid clearance from the wound site(2)(3).

Biomimetic scaffolds provide an innovative solution to this limitation. By mimicking the structure and function of the extracellular matrix, scaffolds act as a supportive framework that enhances exosome stability, prolongs their release, and maintains a moist, pro-healing environment. Recent studies have shown that scaffold–exosome combinations create sustained bioactive signaling at the wound bed, leading to improved granulation tissue formation, re-epithelialisation, and overall wound closure(1)(2).

In this case, the integration of MSC-derived exosomes with a biomimetic scaffold resulted in accelerated healing of a chronic diabetic wound, with more than 70% reduction in wound size within 11 weeks, without complications or adverse effects. This favourable outcome is consistent with preclinical evidence supporting the synergistic effect of scaffolds and exosomes in overcoming the limitations of conventional therapies. Although limited to a single patient, this case highlights the potential of scaffold–exosome therapy as a safe and promising adjunct for chronic wound management, warranting further clinical studies to establish its long-term efficacy and reproducibility.

CONCLUSION

This case demonstrates that the use of a biomimetic scaffold loaded with MSC-derived exosomes can significantly accelerate healing in chronic wounds of uncontrolled diabetic patients. The scaffold provided an extracellular matrix-like structure that supported cell adhesion and proliferation, while the exosomes delivered angiogenic, anti-inflammatory, and regenerative factors directly to the wound site. Findings from previous studies support that exosome-loaded scaffolds enhance the stability and bioavailability of exosomes, modulate inflammation, promote angiogenesis, and stimulate epithelialisation, thereby addressing the limitations of conventional dressings and free exosome applications. This approach represents a promising adjunctive therapy for chronic wound management in diabetic patients, warranting further clinical studies to validate efficacy, optimise scaffold-exosome integration, and evaluate long-term safety.

www.woundconference.com.my